Elder Quest: Enhancing Learning with Video Games

Alice Pomidor, MD, Ken Brummel-Smith, MD, Suzanne Baker, MA

Florida State University College of Medicine, Department of Geriatrics, Tallahassee, Florida

This research was supported by the Donald W. Reynolds Foundation and the Florida State University College of Medicine. The investigators retained full independence in the conduct of this research.

Abstract

Purpose: To evaluate use, acceptance and learning outcomes of an interactive video game aimed at helping medical students learn the AAMC geriatric competencies.

Background: Video games motivate learning with fun interactions providing immediate feedback and reinforcement of concepts. The first module of a video game was completed in 2010, during which the player starts a quest to save the Grey Sage and the Kingdom. A website (www.elderquest.org) hosts the game for play.

Methods: 4th year medical students on their geriatrics rotations during 2011 were recruited for the pilot study. Game content on medication management, delirium, falls, and changes of aging was labeled according to the AAMC competencies. Learner performance data collection measures were built into the ElderQuest (EQ) video game by Brainstorm Rising. An evaluation tool on learner usage and acceptance of EQ was administered upon completion of student participation in the study.

Results: To date, 17 students have chosen to participate as game players and 5 as a comparison group. Gamers were 76.5% female compared to 40% of nongamers. Almost two-thirds of the respondents (11 of 17 or 64.7%) rated EQ as effective or very effective on a 9-point scale for gaining knowledge in medication management, 7 of 15 (46.7%) for delirium, 8 of 16 (50.0%) for changes of aging, and 5 of 15 (33.3%) for falls. 70.6% (12 of 17) agreed or strongly agreed that playing EQ increased their proficiency in geriatrics in addition to what was learned during the clerkship, and 13 of 17 (76.5%) also agreed or strongly agreed that EQ helped them recognize areas in which they need additional knowledge. 13 of 17 (76.5%) agreed that EQ made it more likely that they would remember and apply medical lessons over the long term. 14 of 16 (87.5%) respondents felt that videogames should be used more often to enhance healthcare education. Nongamers cited technical difficulties, lack of gameplay skills and time constraints as their major reasons for nonparticipation. Mean gamers' clerkship exam scores were 79.5% (sd=4.3) compared to 80.0% (sd=5.89) for nongamers, with clerkship final grades showing no significant difference.

Conclusions: Students can use an educational geriatrics video game to reinforce the AAMC competencies content of a geriatrics clerkship. Student acceptance of videogaming as a learning tool was high and believed to improve long-term retention. Refinement of the game is needed to address technical issues and gameplay concerns.

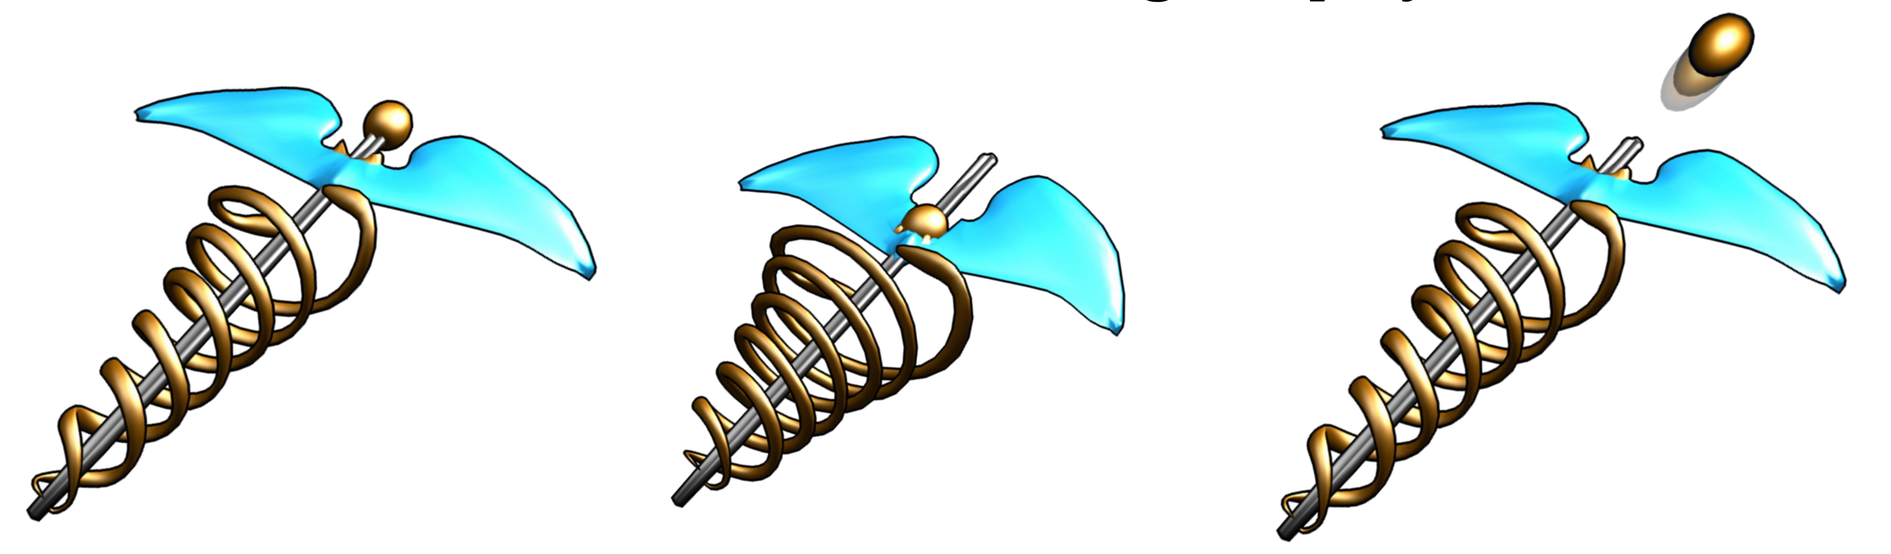

METHODS

Why Video Games?

- New generations of learners have grown up playing video games, and continue to do so as adults.
- Preferred learning strategies of the Millenials/Net generations emphasize technology-based, visually stimulating, interactive presentations.
- Using a game-based approach adds a "coolness" factor to geriatrics education, which is rarely perceived as a cutting-edge field.
- 3-D video games can model geriatric situations and team care with first-person play in a fun, reward-based, self-motivating environment
- Game difficulty and content can be tailored to meet the needs of different levels of learner or health care professions.

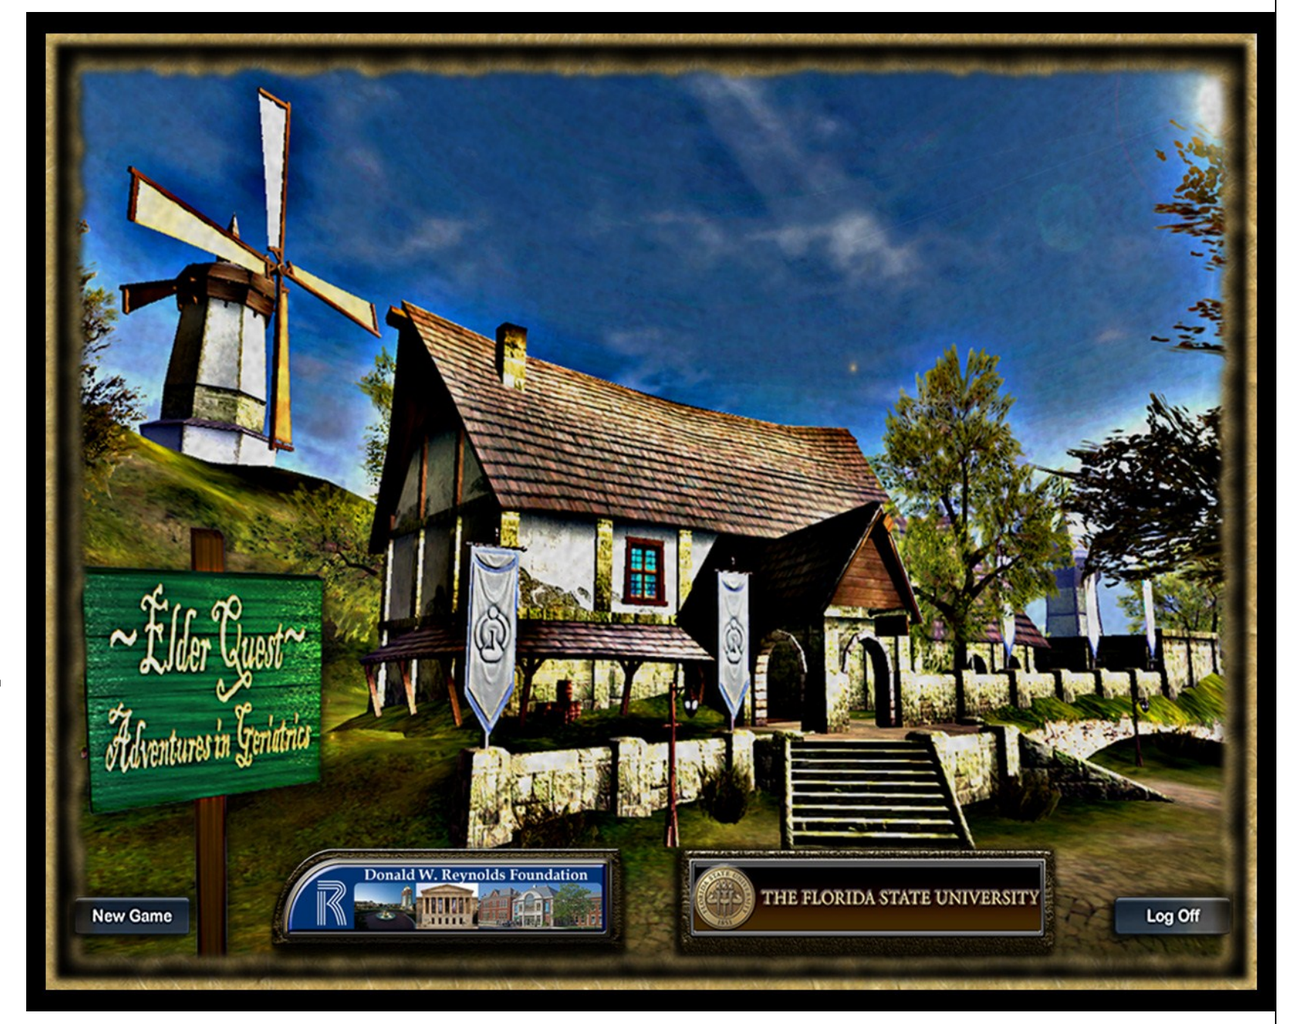

~Elder Quest~
Adventures in Geriatrics
New Game
Donald W. Reynolds Foundation
THE FLORIDA STATE UNIVERSITY
Log Off

AAMC Competencies Applied: Module 1

MEDICATION MANAGEMENT
- Explain impact of age-related changes on drug selection and dose.
- Identify medications that should be avoided or used with caution in older adults and explain the potential problems associated with each.

COGNITIVE AND BEHAVIORAL DISORDERS
- Formulate a differential diagnosis and implement initial evaluation in a patient who exhibits cognitive impairment.
- Urgently initiate a diagnostic work-up to determine the root cause (etiology) of delirium in an older patient.
- Perform and interpret a cognitive assessment in older patients for whom there are concerns regarding memory or function.
- Develop an evaluation and non-pharmacologic management plan for agitated, demented, or delirious patients.

SELF-CARE CAPACITY
- Identify and assess safety risks in the home environment, and make recommendations to mitigate them.

ATYPICAL PRESENTATION OF DISEASE
- Identify physiologic changes of aging for each organ system and their impact on the patient, including their contribution to homeostenosis.

Subjects • Learner Performance • Game Acceptance

1. Subjects
- Year 4 medical students during calendar year 2011
- **112** students on their geriatrics clerkship during 2011 invited to participate in study
- **21** students chose to play **(19%)**
- Age, gender of players vs. overall class similar (26.8 vs. 26.2 yrs.)

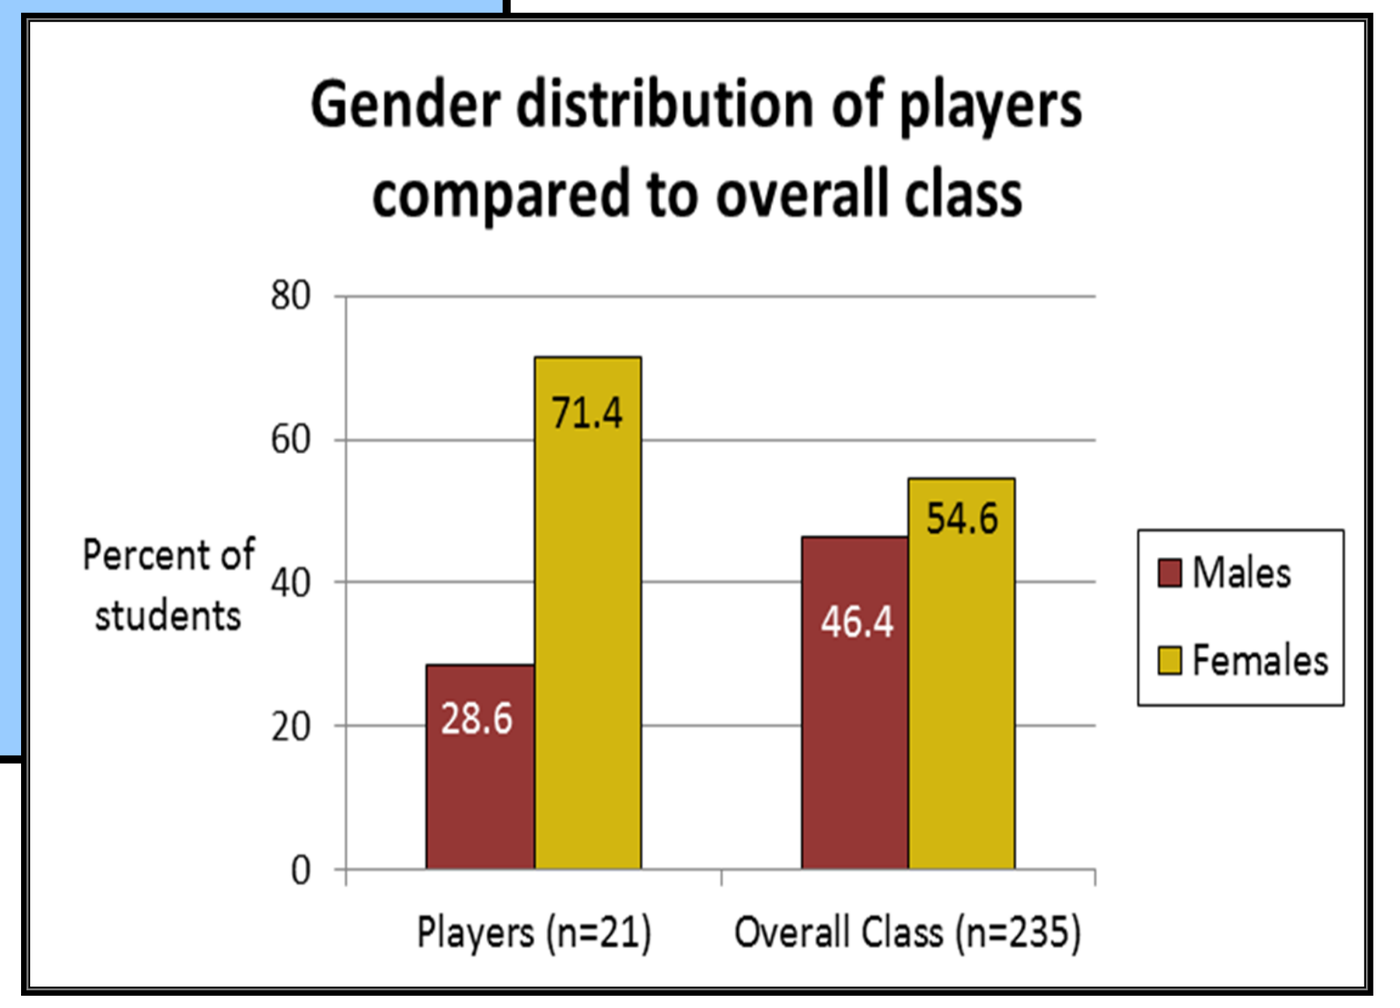

Gender distribution of players compared to overall class
Percent of students
80
60
40
20
0
28.6
71.4
46.4
54.6
Players (n=21)
Overall Class (n=235)
Males
Females

2. Learner Performance Data
- Game content coded to the AAMC competencies
- Performance data collection measures built into game design software by Brainstorm Rising
- In-game debrief screen feedback provided to student at each level

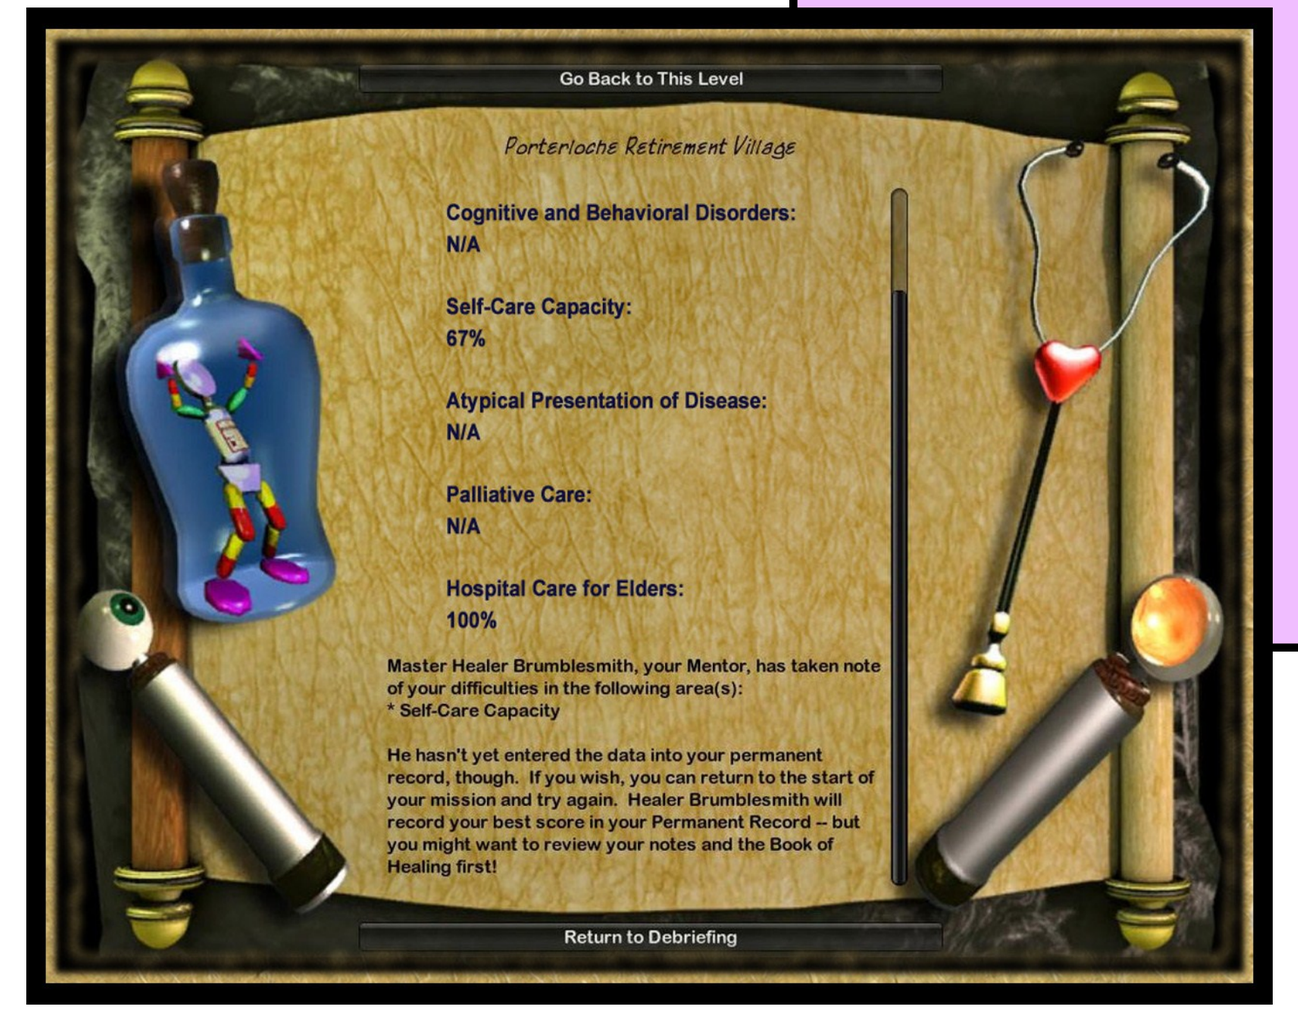

Go Back to This Level
Porterloche Retirement Village
Cognitive and Behavioral Disorders:
N/A
Self-Care Capacity:
67%
Atypical Presentation of Disease:
N/A
Palliative Care:
N/A
Hospital Care for Elders:
100%
Return to Debriefing

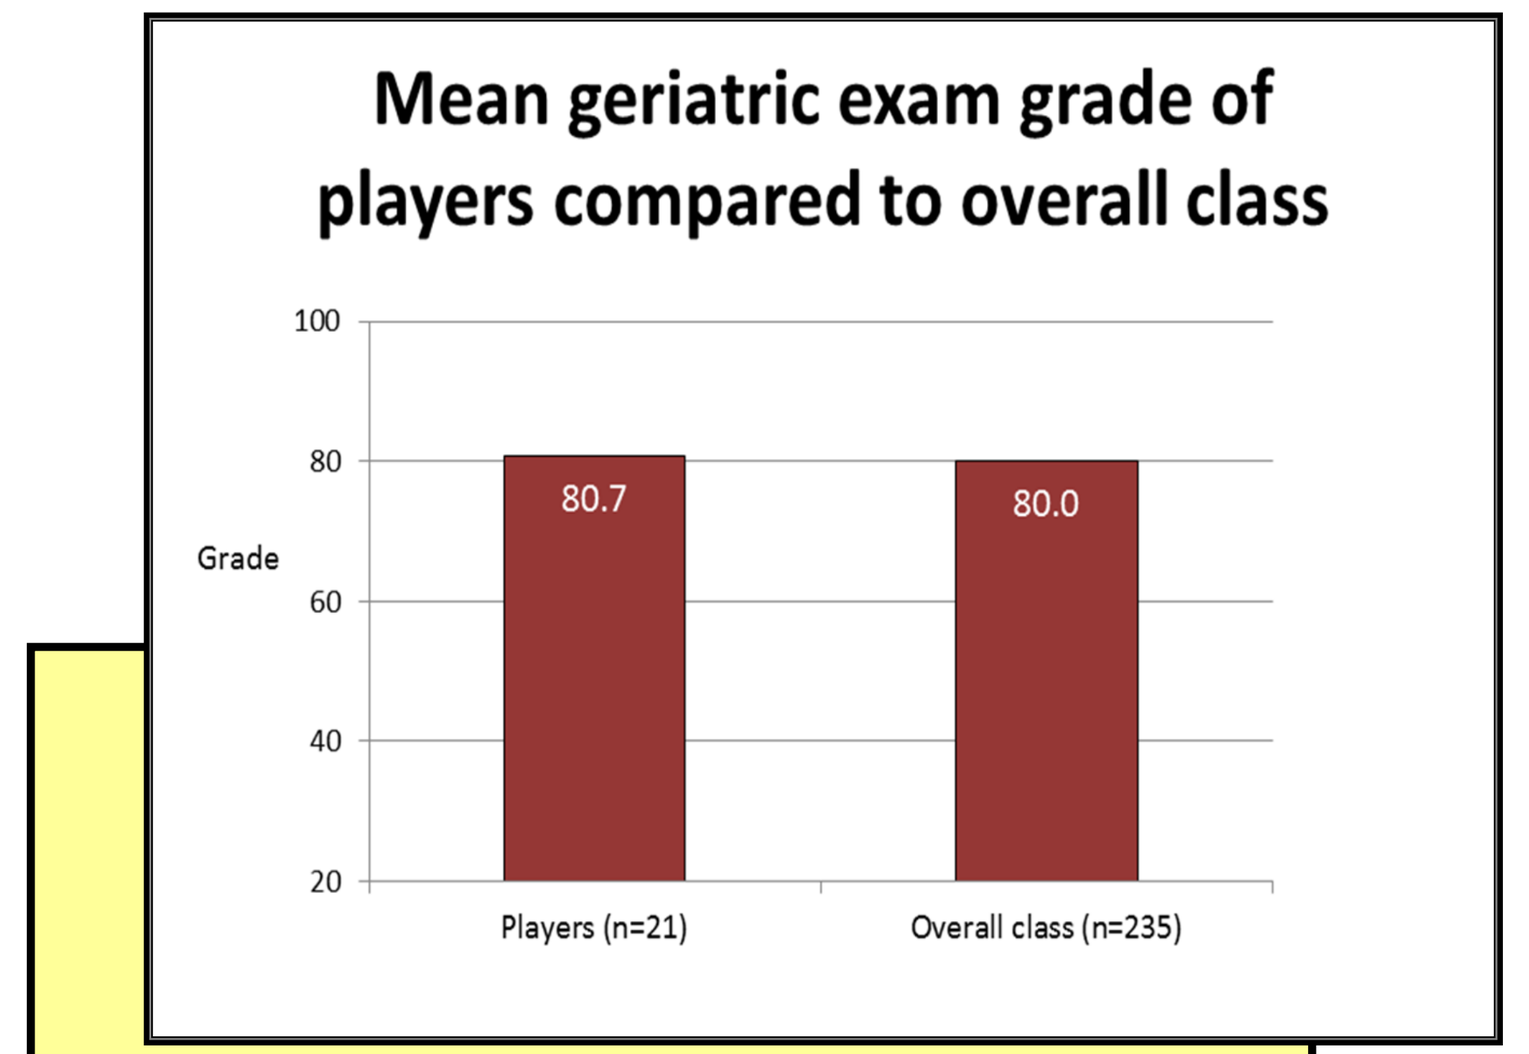

Mean geriatric exam grade of players compared to overall class
Grade
100
80
60
40
20
80.7
80.0
Players (n=21)
Overall class (n=235)

5. Learner Outcomes
- No difference in geriatrics clerkship final exam scores between players and overall class
- No differences in overall clerkship ratings

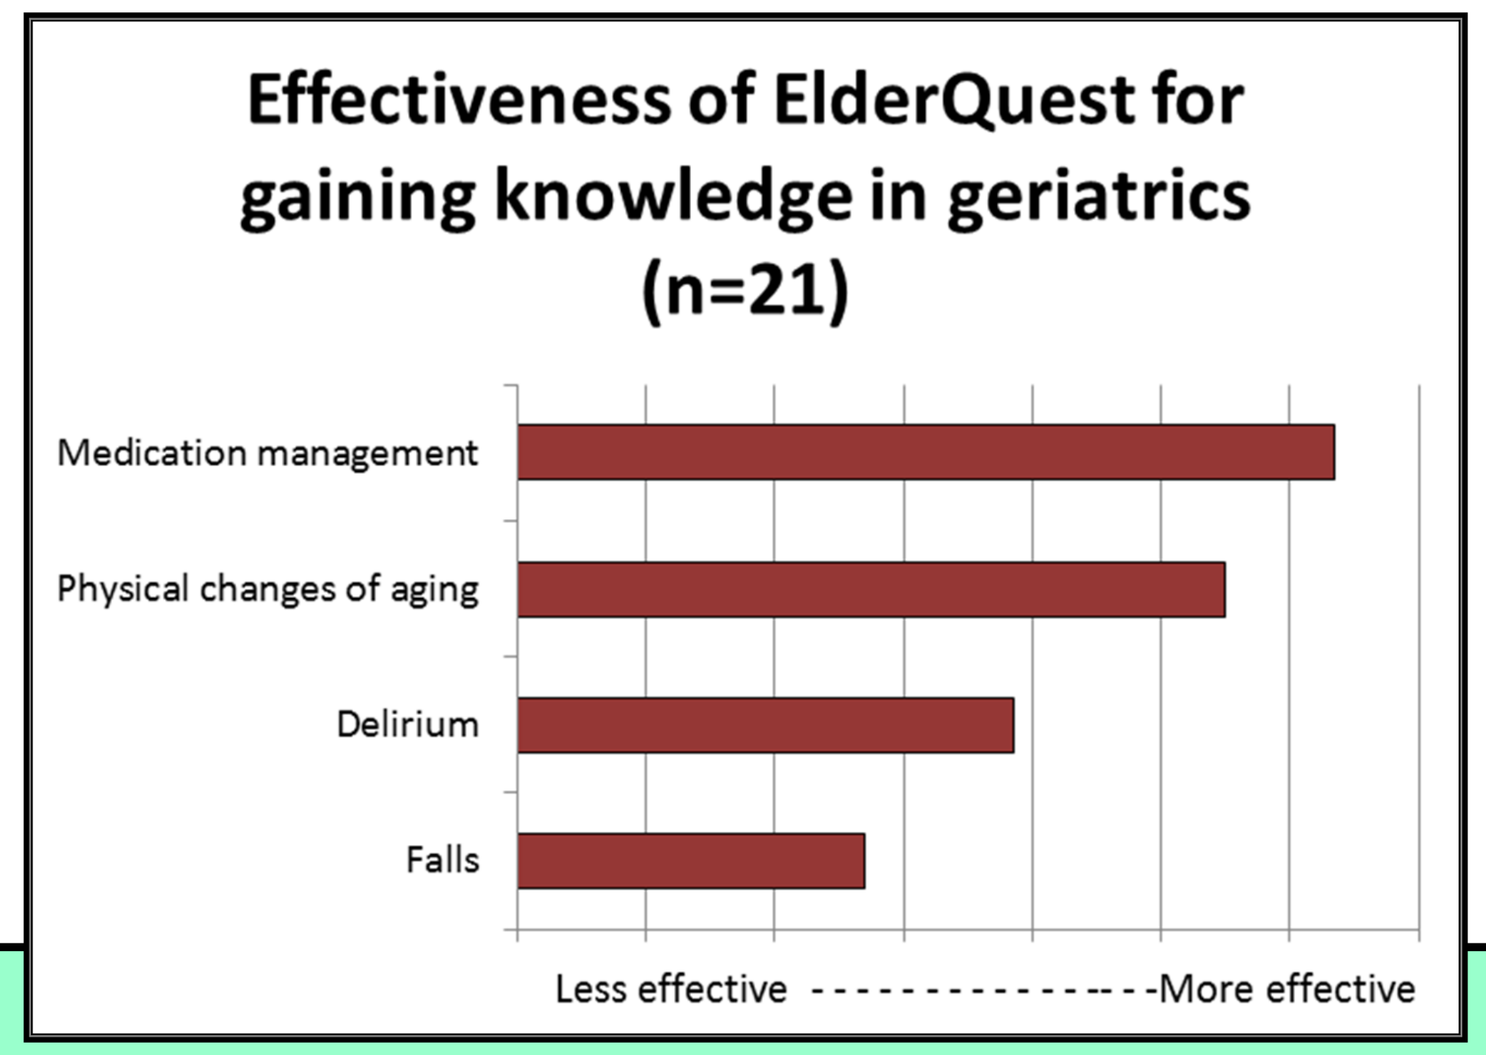

Effectiveness of ElderQuest for gaining knowledge in geriatrics (n=21)
Medication management
Physical changes of aging
Delirium
Falls
Less effective ---------------More effective

4. Effectiveness in Competency Areas
- Students asked to rate effectiveness of *ElderQuest* for gaining knowledge on a Likert scale
- Areas assessed matched to game content:
 - Medication management
 - Delirium
 - Physical changes of aging
 - Falls

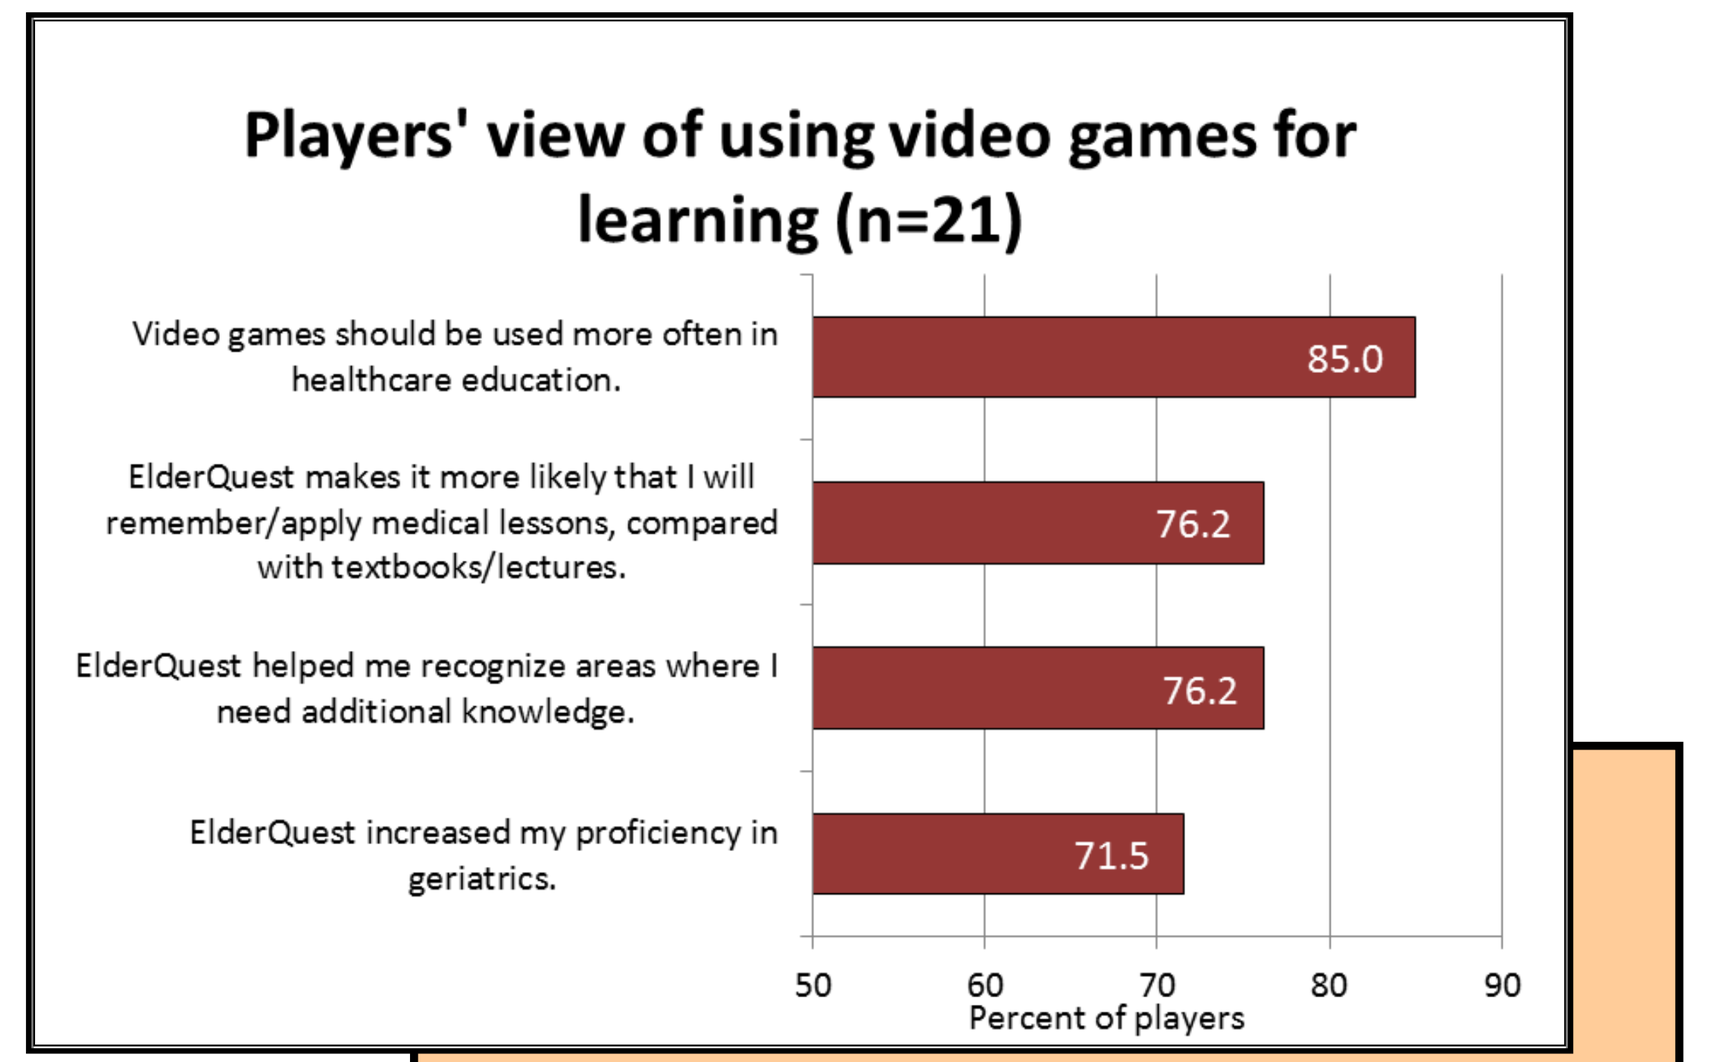

Players' view of using video games for learning (n=21)
Video games should be used more often in healthcare education.
85.0
ElderQuest makes it more likely that I will remember/apply medical lessons, compared with textbooks/lectures.
76.2
ElderQuest helped me recognize areas where I need additional knowledge.
76.2
ElderQuest increased my proficiency in geriatrics.
71.5
50
60
70
80
90
Percent of players

3. Player Satisfaction/ Retention of Learning
- Survey conducted after students played the game
- Rated themselves as more likely to retain learning
- Helped recognition of areas where learning was needed
- Believed to increase proficiency in geriatrics

Game Design Outcomes

6. Blend of medical education and game play was well accepted

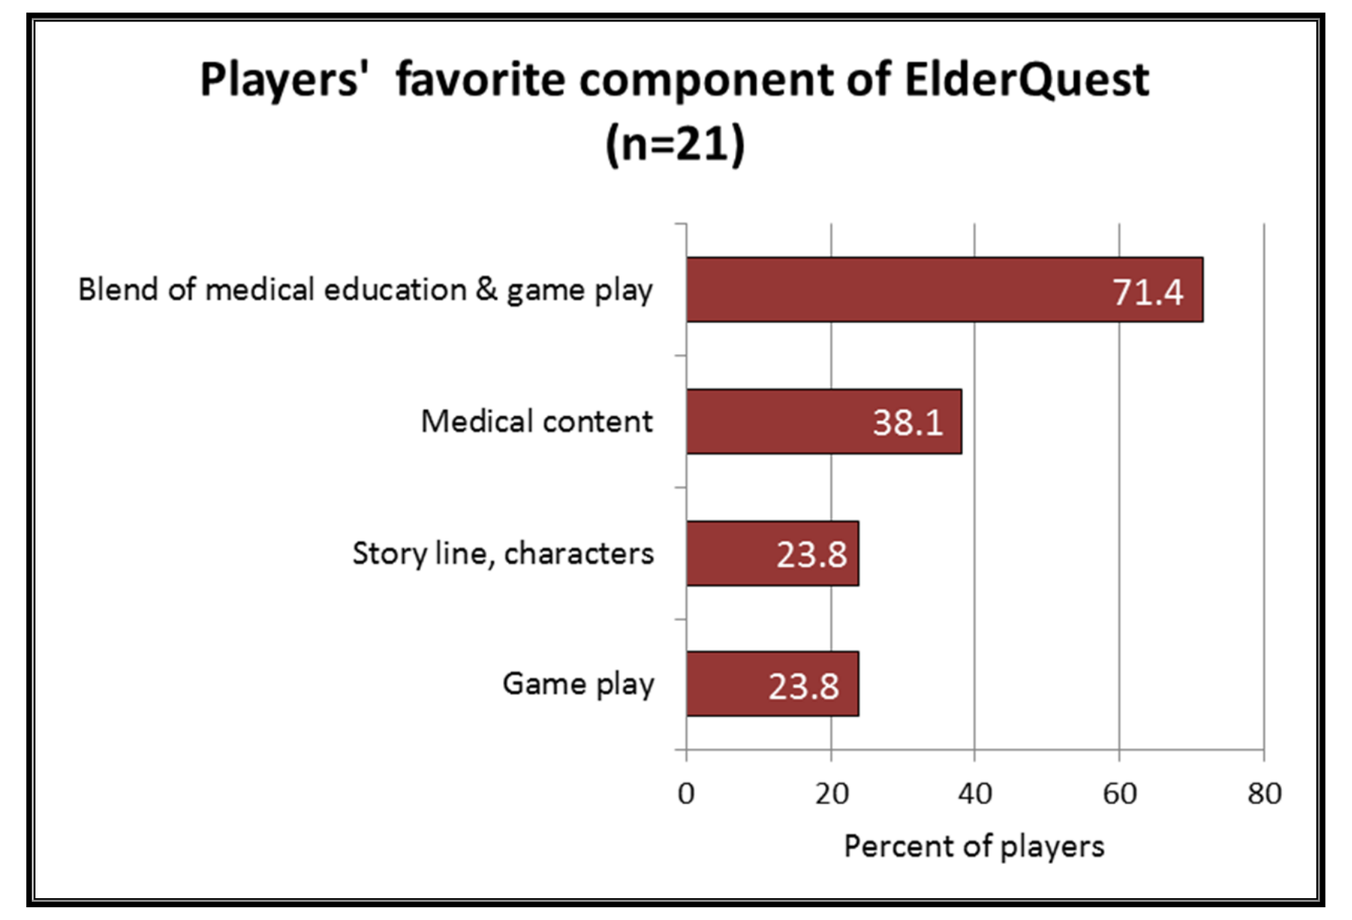

Players' favorite component of ElderQuest (n=21)
Blend of medical education & game play
71.4
Medical content
38.1
Story line, characters
23.8
Game play
23.8
0
20
40
60
80
Percent of players

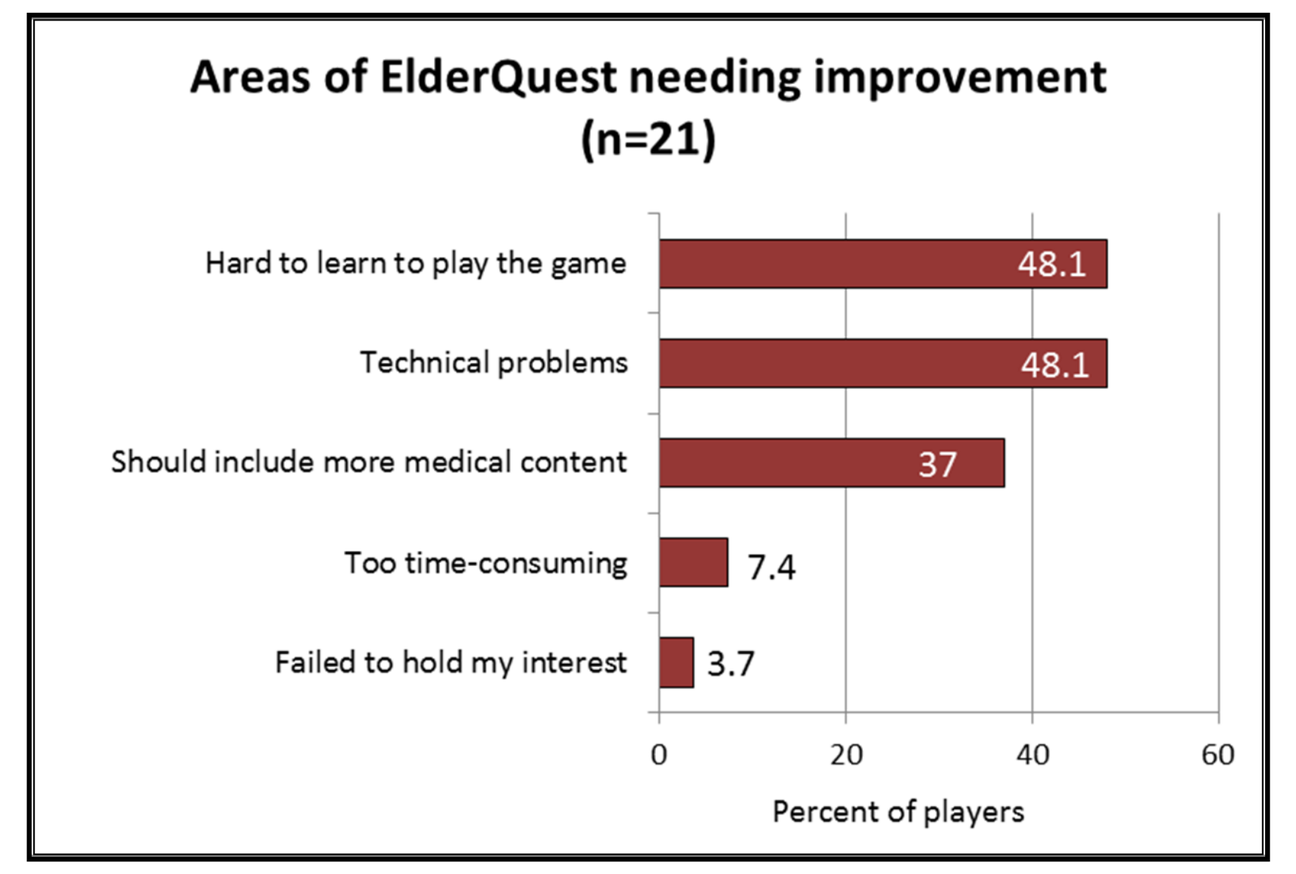

Areas of ElderQuest needing improvement (n=21)
Hard to learn to play the game
48.1
Technical problems
48.1
Should include more medical content
37
Too time-consuming
7.4
Failed to hold my interest
3.7
0
20
40
60
Percent of players

7. Technical issues/game support need improvement

Conclusions • Future Plans

- Students can use an educational geriatrics video game (*ElderQuest*) to reinforce the AAMC competencies content of a geriatrics clerkship.
- Student acceptance of video gaming as a learning tool was high and believed to improve long-term retention.
- Refinement of the game is needed to address technical issues and gameplay concerns.
- Data from the pilot study will guide the ongoing development and refinement of *ElderQuest's* educational content and overall design.
- Technical support and free access (www.elderquest.org) are essential.

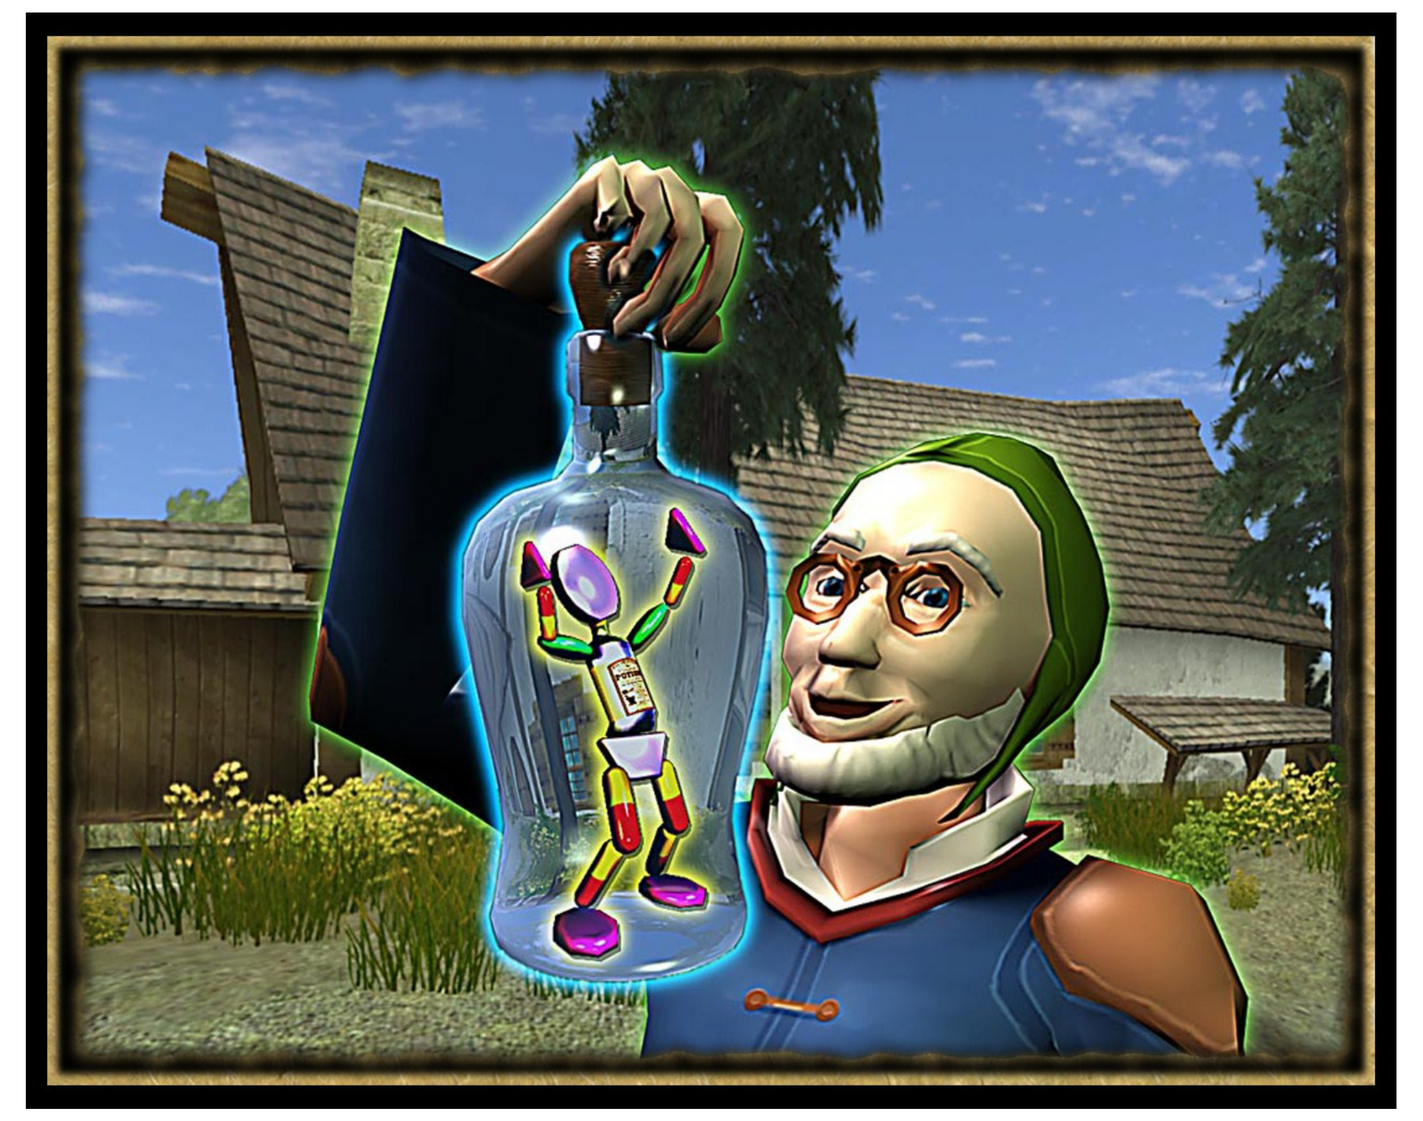